PS03-2265

Psychoactive Effects of GH001 in Patients With Treatment-Resistant Depression: Results From a Phase 2b, Double-Blind, Randomised Controlled Trial

Fabian Devlin[1*], Bernhard T. Baune[2,3,4], Narcís Cardoner[5], Wiesław J. Cubała[6], Kelly Doolin[7], Rosa Maria Dueñas Herrero[8], Matilde Elices[9,10], Katarzyna Jakuszkowiak-Wojten[6], Luboš Janů[11], John R. Kelly[12,13], Rachael MacIsaac[7], Shane J. McInerney[14], Alexander Nawka[15], Tomáš Páleníček[16], Victor Perez Sola[9,10,17,18], Andreas Reif[19,20], Claus Bo Svendsen[7], Claire Sweeney[7], Michael E. Thase[21,22], Velichka Valcheva[7], Eduard Vieta[23,24], Johannes G. Ramaekers[25]

[1]Mental Health Research for Innovation Centre, Mersey Care NHS Foundation Trust, United Kingdom; [2]Department of Psychiatry, University of Muenster, Muenster, Germany; [3]Department of Psychiatry, University of Melbourne, Melbourne, VIC, Australia; [4]The Florey Institute of Neuroscience and Mental Health, Parkville, VIC, Australia; [5]Hospital Santa Creu i Sant Pau, Mental Health Research Group, Institut de Recerca Sant Pau, Universitat Autònoma de Barcelona, CIBERSAM Barcelona, Spain; [6]Department of Psychiatry, Faculty of Medicine, Medical University of Gdańsk, Gdańsk, Poland; [7]GH Research, Dublin, Ireland; [8]Parc Sanitari Sant Joan de Deu Hospital de Dia de Numancia, Barcelona, Spain; [9]Centre for Biomedical Research in Mental Health (CIBERSAM), Madrid, Spain; [10]Mental Health Institute, Hospital del Mar, Barcelona, Spain; [11]A-Shine SRO, Pilsen, Czechia; [12]Department of Psychiatry, Tallaght University Hospital, Dublin, Ireland; [13]Trinity College Dublin and Tallaght University Hospital, Dublin, Ireland; [14]Department of Psychiatry, University of Galway, Galway, Ireland; [15]Institut Neuropsychiatrické Péče, Praha, Czechia; [16]Psyon s.r.o., Prague, Czechia; [17]Neurosciences Research Group, Hospital del Mar Research Institute (IMIM), Barcelona, Spain; [18]Department of Psychiatry and Department of Experimental and Health Sciences, Pompeu Fabra University, Barcelona, Spain; [19]Department of Psychiatry, Psychosomatic Medicine and Psychotherapy, University Hospital Frankfurt – Goethe University, Frankfurt am Main, Germany; [20]Fraunhofer Institute for Translational Medicine and Pharmacology ITMP, Theodor-Stern-Kai 7, 60596 Frankfurt am Main, Germany; [21]Department of Psychiatry, University of Pennsylvania, Philadelphia, PA, USA; [22]Corporal Michael J. Crescenz Veterans Affairs Medical Center, Philadelphia, PA, USA; [23]Hospital Clinic de Barcelona, Institute of Neuroscience, University of Barcelona, IDIBAPS, CIBERSAM, Barcelona, Catalonia, Spain; [24]Centre for Biomedical Research in Mental Health (CIBERSAM), Madrid, Spain; [25]Faculty of Psychology and Neuroscience, Maastricht University, Maastricht, Netherlands.

*Presenting author: Fabian Devlin; fabiandevlin@hotmail.com

Background

- GH001 is a synthetic form of mebufotenin (5-methoxy-N,N-dimethyltryptamine [5-MeO-DMT]) developed for pulmonary inhalation which has shown potential to treat treatment-resistant depression (TRD)[1]
- Previous trials suggest that administration of GH001 elicits strong psychoactive effects (PsE) in healthy volunteers and in patients with TRD[1,2]
- Recent evidence suggests that the intensity of the acute psychoactive experience may be a predictor of clinical response for psychedelic agents in the treatment of psychiatric disorders [3-5]
- Numerous questionnaires including the Mystical Experiences Questionnaire (30 Questions [MEQ30]) and the Challenging Experience Questionnaire (CEQ) exist for quantifying the PsE[2]; however, these questionnaires are time-consuming to administer and not specific to the PsE of mebufotenin
- The proprietary Peak Experience Scale (PES) was developed to rapidly assess PsE after GH001 administration

Objective

- To describe the PsE of GH001 in the double-blind part of a Phase 2b trial in patients with TRD, as evaluated by the PES, MEQ30, and CEQ

Methods

Trial Design

- In the double-blind part of this Phase 2b multicentre trial (NCT05800860), patients with TRD were randomised 1:1 to receive an individualised dosing regimen (IDR) of up to three escalating doses of GH001 (6, 12, and 18 mg) or placebo IDR on a single day (1-hour interval between doses) (Figure 1)
- The trial was conducted under the supervision of qualified healthcare professionals, providing psychological support per standard-of-care, but without any planned psychotherapeutic intervention before, during, or after dosing

Assessments

- PsE were assessed via the PES, which measures three core features of the PsE (intensity, profoundness, and perceived loss of control), each on a visual analogue scale ranging from 0 to 100, with higher scores indicating stronger PsE (Figure 2)
- A peak experience (PE) was defined as an average total score of ≥75 across the three subscores
- The MEQ30 was administered to assess mystical-type experiences, and the CEQ was administered to characterise and quantify any potentially challenging experiences
- The three PsE assessments were assessed once the PsE had subsided
- Duration of the PsE, defined as the time from study drug administration to the time when the PsE had subsided, was reported by the investigator

Figure 1. Trial Design

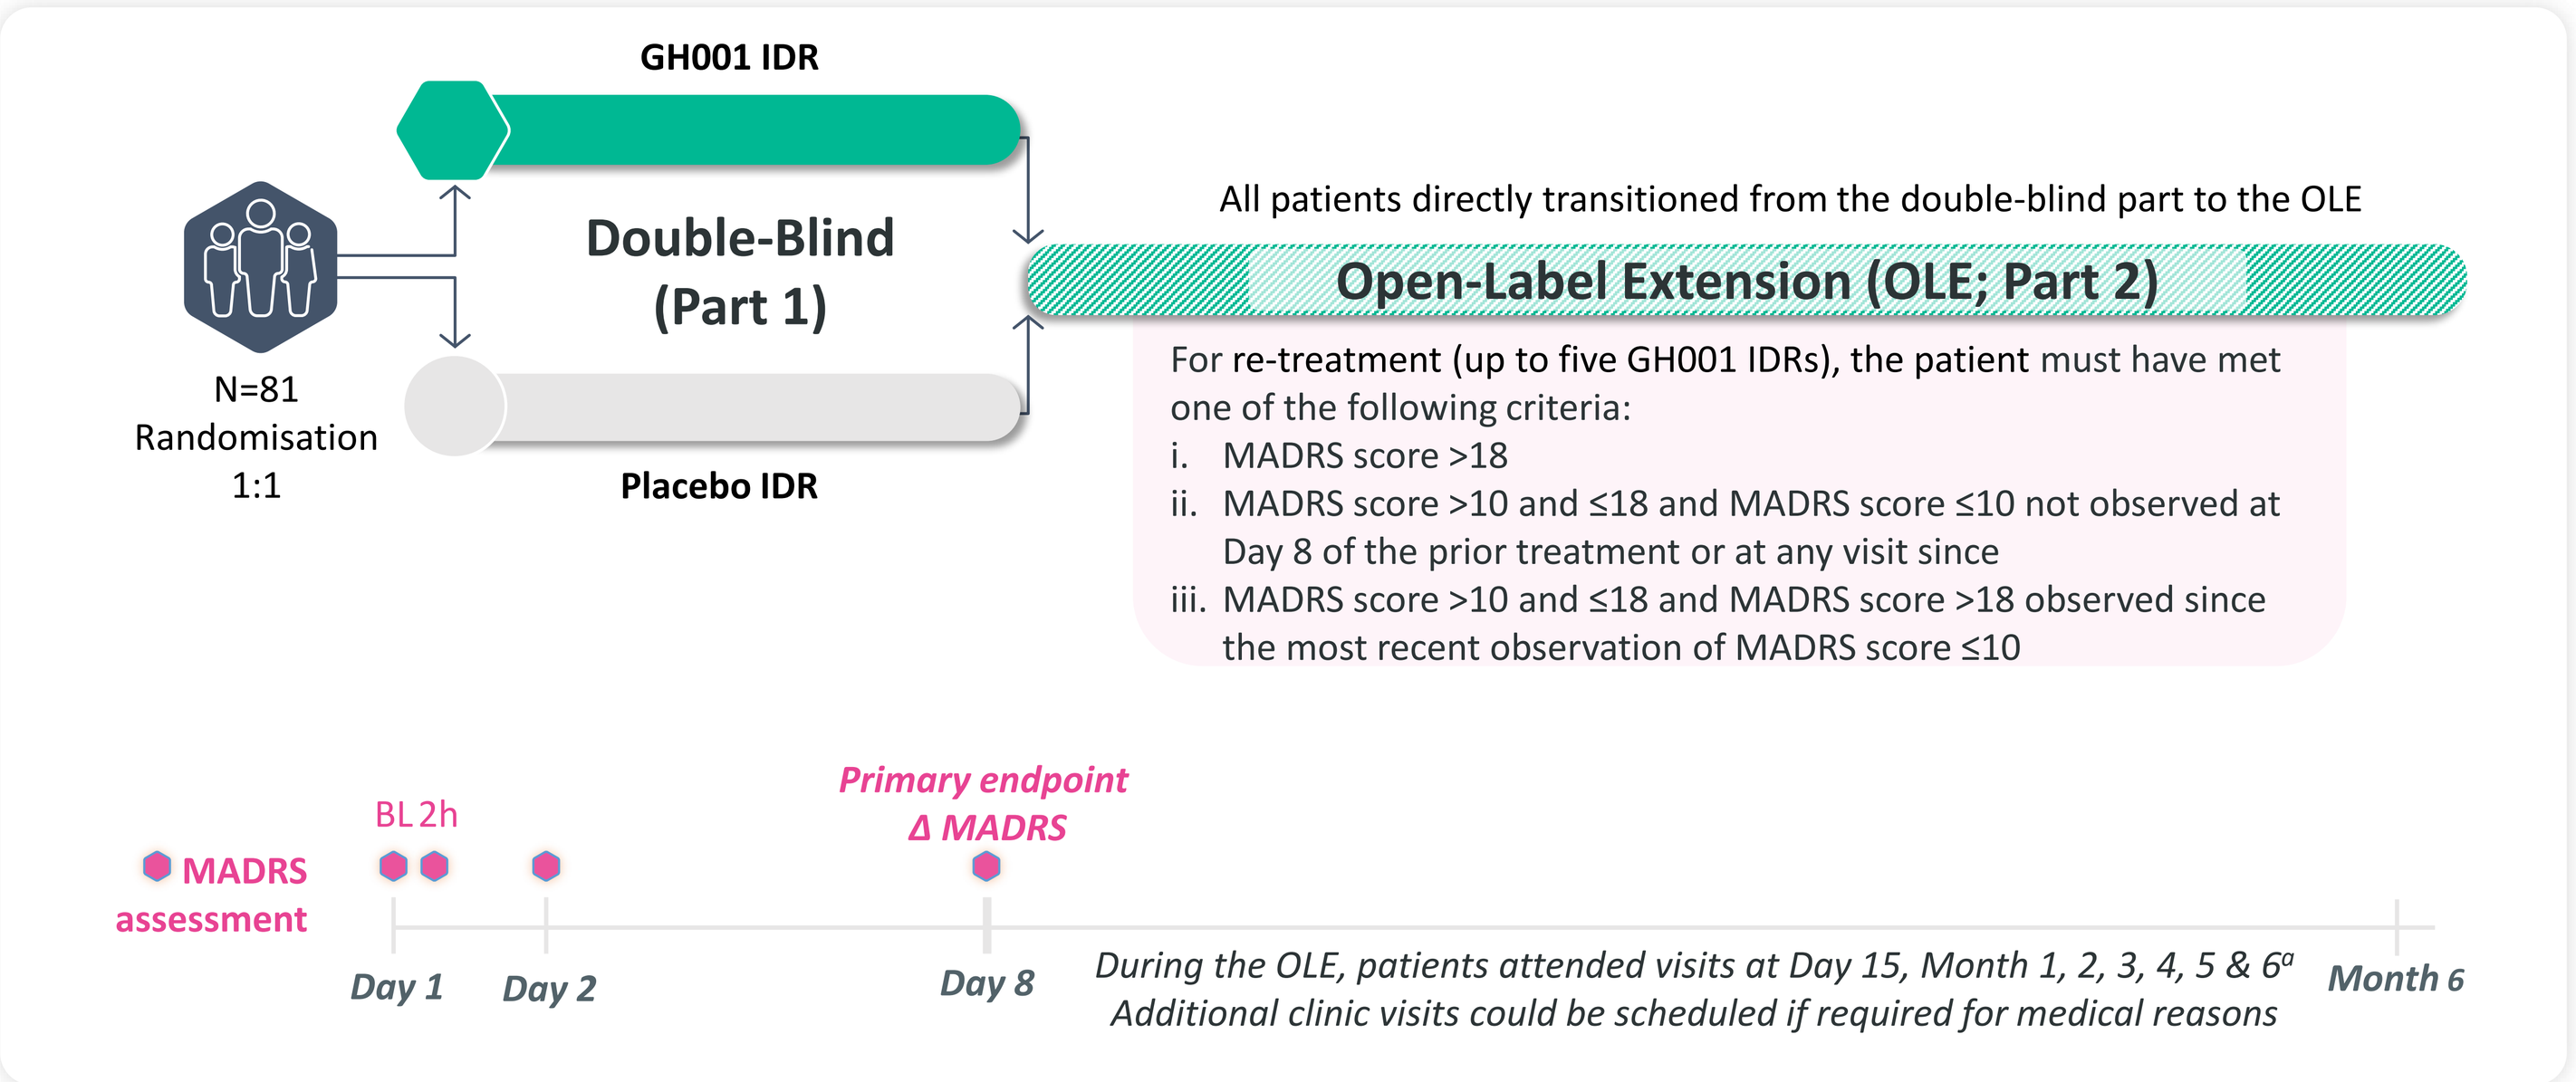
GH001 IDR
Double-Blind
(Part 1)
N=81
Randomisation
1:1
Placebo IDR
All patients directly transitioned from the double-blind part to the OLE
Open-Label Extension (OLE; Part 2)
For re-treatment (up to five GH001 IDRs), the patient must have met one of the following criteria:
i. MADRS score >18
ii. MADRS score >10 and ≤18 and MADRS score ≤10 not observed at Day 8 of the prior treatment or at any visit since
iii. MADRS score >10 and ≤18 and MADRS score >18 observed since the most recent observation of MADRS score ≤10
MADRS assessment
BL 2h
Primary endpoint
Δ MADRS
Day 1
Day 2
Day 8
During the OLE, patients attended visits at Day 15, Month 1, 2, 3, 4, 5 & 6a
Additional clinic visits could be scheduled if required for medical reasons
Month 6

a. Patients also attended assessment visits on Day 2 (phone call) and Day 8 after each re-treatment.
Abbreviations: BL = Baseline; h = Hour; IDR = Individualised dosing regimen; MADRS = Montgomery–Åsberg Depression Rating Scale.

Figure 2. Peak Experience Scale Key Components

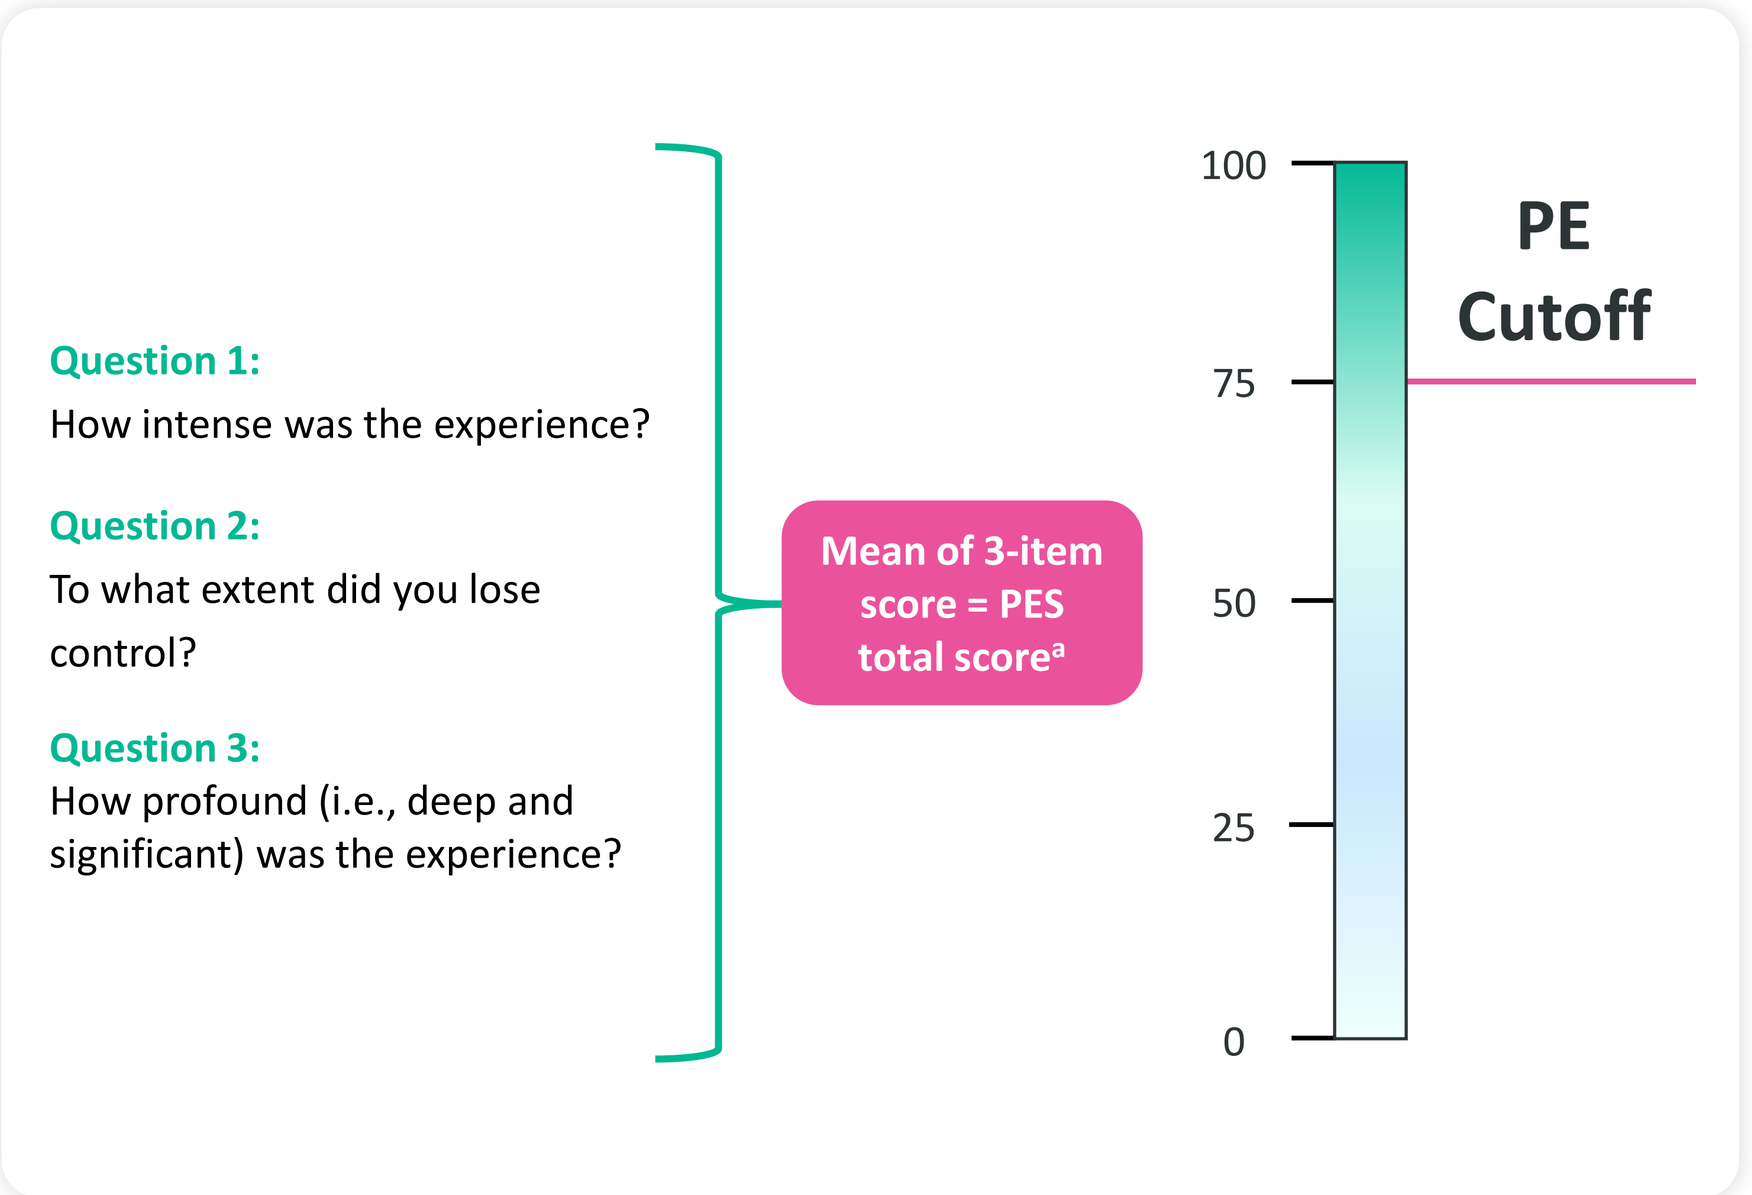
Question 1:
How intense was the experience?
Question 2:
To what extent did you lose control?
Question 3:
How profound (i.e., deep and significant) was the experience?
Mean of 3-item score = PES total scorea
100
75
50
25
0
PE Cutoff

a. Patients with a PES total score of ≥75 were deemed to have achieved a peak experience.
Abbreviations: PE = Peak experience; PES = Peak experiences scale.

Results

- In the double-blind part of this trial, 81 patients were randomised to receive a GH001 IDR (n=40) or placebo IDR (n=41)
 - The mean (SD) age was similar in both groups (41.6 [11.4] and 43.9 [10.9] in the GH001 and placebo group, respectively), and most patients were female and had not previously used any psychedelic

Table 1. Baseline Demographics and Clinical Characteristics

	GH001 (N=40)	Placebo (N=41)
Patient Disposition		
Completed double-blind part, n (%)	40 (100)	41 (100)
Demographics		
Age (years), mean (SD)	41.6 (11.4)	43.9 (10.9)
Female, n (%)	24 (60.0)	22 (53.7)
Race, White, n (%)	40 (100)	41 (100)
BMI (kg/m^2) at screening, mean (SD)	24.8 (4.3)	27.5 (6.3)
Previously used any psychedelic (lifetime), n (%)	4 (10.0)	5 (12.2)
Baseline Disease Characteristics		
MADRS Total Score, mean (SD)	29.0 (5.4)	28.2 (4.6)
HAM-D-17 Total Score, mean (SD)	24.9 (2.7)	24.6 (2.3)
Maximum Dose in IDR[a]		
6 mg GH001 or one placebo dose, n (%)	9 (22.5)	0 (0)
6+12 mg GH001 or two placebo doses, n (%)	21 (52.5)	0 (0)
6+12+18 mg GH001 or three placebo doses, n (%)	10 (25.0)	41 (100)

[a]Up to three doses of GH001 or placebo were administered to each patient.
Abbreviations: BMI = Body mass index; HAM-D-17 = Hamilton Depression Rating Scale; IDR = Individualised dosing regimen; MADRS = Montgomery-Åsberg Depression Rating Scale; SD = Standard deviation.

- Overall, marked PsE were observed in patients receiving GH001 while minimal effects were observed in those receiving placebo
- Investigator-reported median durations of PsE in patients who received GH001 were 9, 14 and 11.5 minutes for the 6, 12 and 18 mg doses respectively, and the median duration did not appear to be dose dependent
- In the placebo group, the median PsE duration was 0 for the first, second and third doses and ranged between 0 and 15 minutes
- Total PES scores increased after each subsequent GH001 dose, with median scores per individual dose of 46.7, 85.0, and 95.7 for the 6, 12, and 18 mg doses, respectively (Table 2); PES scores were markedly lower in patients who received placebo (range, 0–20.7)
- Overall, 35 of 40 patients (87.5%) in the GH001 group and no patients in the placebo group achieved a PE at their maximum individual dose level, with 20.0, 61.3, and 80.0% achieving a PE at the 6, 12, and 18 mg GH001 doses, respectively
- In patients who received GH001 and complied with dose escalation per the IDR, 94.6% (35/37) achieved a PE at their maximum individual dose level
- There was no clear dose-related increase in MEQ30 total scores by individual dose with median scores of 1.0, 1.4, and 1.4 after the 6, 12, and 18 mg doses, respectively. In the placebo group, the median MEQ30 total score was zero and ranged from 0 to 1.10
- In the GH001 group, CEQ scores were low, suggesting that the acute PsE were well tolerated in the majority of patients. In the placebo group, the median CEQ total score was zero, ranging between 0 and 0.6

Table 2. Psychoactive Effect Assessment (by Individual Dose)

Assessment	GH001 6 mg	GH001 12 mg	GH001 18 mg	PBO Dose 1	PBO Dose 2	PBO Dose 3
N	40	31	10	41	41	41
Peak Experience Scale[a]						
Total score, median (range)	46.7 (1.7, 100)	85.0 (35.3, 100)	95.7 (1, 100)	0.7 (0, 5)	0.7 (0, 11)	0.7 (0, 20.7)
PE achieved, n (%)	8 (20.0)	19 (61.3)	8 (80.0)	0	0	0
Mystical Experience Questionnaire-30[b]						
Total score, median (range)	1.0 (0, 4.2)	1.4 (0.4, 4.4)	1.4 (0, 4.1)	0 (0, 0.1)	0 (0, 1.1)	0 (0, 1.1)
Challenging Experience Questionnaire[c]						
Total score, median (range)	0.10 (0, 0.6)	0.15 (0, 0.6)	0.24 (0, 0.6)	0 (0, 0.5)	0 (0, 0.6)	0 (0, 0.6)

[a]The PES score comprised of a visual analogue scale (scored 0-100), with a total score ≥75 qualifying as a peak experience. [b]The MEQ total score is the mean weighted average score across the four factor scores and reported as a percentage of the maximum possible score. [c]The CEQ total score consists of the average of all transformed questionnaire item scores computed to obtain the total CEQ score (range 0-1).
Abbreviations: PBO = Placebo; PE = Peak experience.

Conclusions

- Administration of GH001 elicited pronounced PsE, as assessed by the PES
- CEQ scores were low suggesting that the acute PsE were well tolerated in the majority of patients
- The brevity and tolerability of the PsE with GH001, coupled with rapid symptom improvement and a favourable safety profile[1], could enhance treatment tolerability and align more closely with patient and provider preferences for scalable clinical use
- The PsE of GH001 will be further evaluated in the open-label extension, which allows for up to five re-treatments with GH001 over a 6-month period

References

1. Reckweg JT, et al. *Front Psychiatry*. 2023;14:1133414.
2. Reckweg J, et al. *Front Pharmacology*. 2021;12:760671.
3. Romeo, B., et al. *J Psychiatr Res.* 2021; 137, 273-282.
4. Ko, K. et al.. *Front. Psychiatry*. 2022; 13, 917199.
5. Romeo, B et al. *Neurosci. Biobehav Rev*. 2025; 172, 106086.

Acknowledgments

This trial was sponsored by GH Research. The sponsor would like to thank the participants in the trial. The sponsor would also like to thank the investigators who conducted this trial. Under the guidance of authors, medical writing and editorial support were provided by Erik O'Hanlon of GH Research Ireland Limited.

Disclosures

FD: Former employee – GH Research; Advisory board – GH Research, **BTB**: Consultant – National Health and Medical Research Council (Australia). Honoraria – Angelini, AstraZeneca, Biogen, BMS, Boehringer Ingelheim, Johnson & Johnson, LivaNova, Lundbeck, Medscape, Otsuka, Pfizer, Roche, Servier, Sumitomo Pharma, Sunovion, Teva, Viatris and Wyeth. Advisory boards – Biogen, Boehringer Ingelheim, Janssen-Cilag, LivaNova, Lundbeck, Medscape, Novartis, GH Research, Otsuka, and Teva. Research grants from private industries or nonprofit funds – AstraZeneca, BMBF (Germany), BMG (Germany), DFG (Germany), ERA PerMed, Fay Fuller Foundation, Horizon Europe (European Union), James & Diana Ramsay Foundation (Adelaide), Johnson & Johnson, Lundbeck, La Marató de TV3, National Health and Medical Research Council (Australia), Sanofi-Synthélabo, and Wellcome Trust (UK), **NC:** Grants – Spanish Ministry of Health, Spanish Ministry of Science and Innovation (CIBERSAM), Strategic Plan for Health Research and Innovation (PERIS) 2016–2020, Recercaixa, and La Marató de TV3. Honoraria – Adamed, Elsevier, Exeltis, Janssen, Lundbeck, Pfizer, and Servier. Advisory Boards – Angelini, Esteve, Janssen, Lundbeck, Novartis, Pfizer, and Viatris. Lectures/Meetings – Janssen, Lundbeck, and Pfizer, **WJC:** Grants – Acadia, Angelini, Beckley Psytech, GH Research, HMNC Brain Health, Intra-Cellular Therapies, Janssen, MSD, Neumora, Novartis, Otsuka, Recognify Life Sciences. Honoraria – Angelini, GH Research, Janssen, and Novartis.Advisory boards – Douglas Pharmaceuticals, GH Research, Janssen, MSD, and Novartis (relationships reported within the last three years), **KD, RM, CS,** and **VV:** Employees and stock option holders of GH Research, **RMDH:** Principal investigator – Beckley Psytech and GH Research. Subinvestigator – Compass, **ME:** Research support – GH Research, Compass, Beckley Psytech, CaixaResearch, La Marató de TV3, and Spanish Ministry of Health, **KJW:** Research support – Alkermes, Allergan, Auspex Pharmaceuticals, Beckley Psytech, Biogen, Cephalon, Compass, Cybin, GH Research, GW Pharmaceuticals, Janssen, KCR, Lilly, Lundbeck, MindMed, Minerva, Novartis, Orion, Otsuka, and Servier, **LJ:** Principal investigator – GH Research, **JRK:** Principal investigator – Compass, GH Research, and Transcend Therapeutics. Consultant – Clerkenwell Health. Grant funding – Health Research Board (ILP-POR-2022-030, DIFA-2023-005,KTA-2024-002), **SJM:** Principal investigator – GH Research and Transcend Therapeutics. Honoraria – Janssen and Lundbeck, **AN:** Principal investigator – GH Research, **TP:** Principal investigator – Compass, GH Research, MAPS,and Ketabon. Shares Psychedelická klinika s.r.o., Spolenčnost pro podporuneurovědního výzkumu s.r.o., and AVI-X Aviation Experts s.r.o. Founder – PSYRES (Psychedelic Research Foundation). Consultant – CB21 Pharma and GH Research, **VPS:** Grants and consultant, advisor, or CME speaker – Almirall, AstraZeneca, Eli Lilly, GlaxoSmithKline, Ferrer, Johnson & Johnson, Lundbeck, Medtronic, Merck, Otsuka, Pfizer, Servier, and the Spanish Ministry of Science and Innovation, **AR:** Honoraria for lectures and/or advisory boards – AbbVie, Boehringer Ingelheim, Cyclerion, Compass, GH Research, Janssen, LivaNova, Medice, MSD, Newron, Sage/Biogen, and Shire/Takeda. Research grants – Medice and Janssen, **CBS:** Consultant and shareholder – GH Research, **MET:** Grants – Acadia, Alkermes, Axsome, Intra-Cellular Therapies, Janssen, National Institute of Mental Health, Otsuka, Patient-Centered Outcomes Research Institute (PCORI), and Takeda. Advisory Boards – Autobahn Therapeutics, Axsome, Clexio Biosciences, Gerson Lehrman Group, GH Research, Lundbeck, Janssen, Johnson & Johnson, Luye Pharma, Merck, Object Pharma, Otsuka, Pfizer, Sage, Seelos Therapeutics, Sunovion, and Takeda. Royalties – American Psychiatric Association Foundation, Guilford Publications, Herald House, Wolters Kluwer, and W Luyi Norton & Company, **EV:** Grants – AB-Biotics, AbbVie, Almirall, AstraZeneca, Boehringer Ingelheim, Bristol Myers Squibb, Celon, Cephalon, Dainippon Sumitomo Pharma, Elan, Ferrer, GH Research, GlaxoSmithKline, Janssen, Lilly, Lundbeck, Orion, Otsuka, Pfizer, Sanofi Aventis, Servier, Sunovion, and Takeda. Honoraria – Abbott, AbbVie, Angelini, AstraZeneca, Bristol Myers Squibb, Cambridge University Press, Elsevier, Farmindustria, Ferrer, Galenica, GlaxoSmithKline, Janssen, Johnson & Johnson, Lilly, Lundbeck, Oxford University Press, Otsuka, Pfizer, Sanofi Aventis, and Viatris. Advisory boards – AbbVie, Angelini, AstraZeneca, Biogen, Biohaven, Bristol Myers Squibb, Celon, Compass, Ferrer, GH Research, Gedeon Richter, HMNC, Idorsia, Janssen, Johnson & Johnson, Jazz, Lilly, Lundbeck, Merck Sharp & Dohme, Novartis, Organon, Otsuka, Pfizer, Roche, Sage, Sanofi Aventis, Servier, Shire, Sunovion, Takeda, and Teva, **JGR:** Scientific advisor – GH Research.

Presented at the European College of Neuropsychopharmacology
38th ECNP Congress, Amsterdam, The Netherlands
October 11 - 14, 2025